ABSTRACT
0048

Non-clinical pharmacokinetics of XMT-1522, a HER2 targeting auristatin-based antibody drug conjugate

Donald A. Bergstrom, Dmitry Gumerov, Natalya Bodyak, Alex Yurkovetskiy, Michael DeVit, Mao Yin, Laura Poling, Joshua D. Thomas, Dongmei Xiao, Elena Ter-Ovanesyan, Charlie Bu, LiuLiang Qin, Alex Uttard, Alex Johnson, Timothy B. Lowinger.
Mersana Therapeutics Inc., Cambridge, MA

Abstract

The ADC XMT-1522 consists of a human IgG1 anti-HER2 monoclonal antibody and the novel, auristatin-based cytotoxic payload XMT-1267 (Auristatin F- hydroxypropylamide). XMT-1522 is being developed for the treatment of patients with HER2-positive cancers, as well as for patients with HER2-expressing tumors not meeting the current diagnostic definition of HER2-positive. The DMPK properties of XMT-1522 have been characterized in vitro in animal plasma and microsomal stability studies and in vivo in plasma and tissue disposition studies and elimination studies, which included biliary, renal and gastric excretion studies. Biological sample analysis for small molecule drug related products and conjugated drug was performed by ESI LC/MS/MS methods and analysis for total antibody was performed by ELISA. The studies confirm that the small molecule XMT-1267, postulated as an active component of XMT-1522, is a primary ADC drug release product. XMT-1267 is further metabolized via i) deamidation of the C-terminal amino group resulting in formation of compound XMT-1521 (auristatin F), the most abundant XMT-1267 metabolite, and ii) demethylation of the N-terminus to produce monomethyl auristatin F-HPA (MMAF-HPA). Both XMT-1521 and MMAF-HPA retain specific anti-tubulin activity. The pharmacokinetic profiles of XMT-1522 have been evaluated in mouse, rat and cynomolgus monkey. In all species XMT-1522 PK was mostly linear, approximately dose proportional, and characterized by extended exposure to conjugated XMT-1267 drug payload. Exposure to free XMT-1267 and XMT-1521 was less than 1/1000th exposure of total XMT-1267. Clearance and volume of distribution were similar for conjugated XMT-1267 and XMT-1519 antibody. These data indicate the vast majority of XMT-1267 in plasma is antibody-conjugated, indicating high stability of the drug polymer linkage in the systemic circulation. XMT-1522 tissue disposition was studied in NCI-N87 xenograft tumor bearing mice. Tissue analysis indicated that both XMT-1267 and its metabolite XMT-1521 were generated intracellularly from XMT-1522, and that the carboxylate-containing active metabolite XMT-1521 was retained in tumor tissue over 2 weeks, suggesting intracellular trapping. XMT-1522 excretion studies, conducted in rat, indicated that the XMT-1267 payload was mainly excreted by the gastrointestinal route. In the first 96 hours after administration 33% of the XMT-1267 dose was excreted in feces, compared to 3% excreted in urine. The major contributing metabolites both in feces and urine were conjugated XMT-1267, XMT-1521, and free XMT-1267. In conclusion, XMT-1522 ADC has plasma kinetics, tissue distribution and excretion profile favorable for clinical evaluation and development.

Introduction XMT-1522 Structure

XMT-1522 Key features:

- Average of ~12 auristatin-derived payload molecules per antibody
- Drug-like properties enabled via Fleximer polymer conjugation
- Built on novel mAb (XMT-1519) optimized for ADC; binds to a unique epitope distinct from trastuzumab or pertuzumab
- Molecular weight of each polymer is ~8 to ~14 kDa by SEC
- 4-5 Auristatin F-HPA payload molecules loaded per polymer
- Intracellular cleavage of drug-polymer linker
- 3-5 polymers conjugated primarily to hinge region cysteines

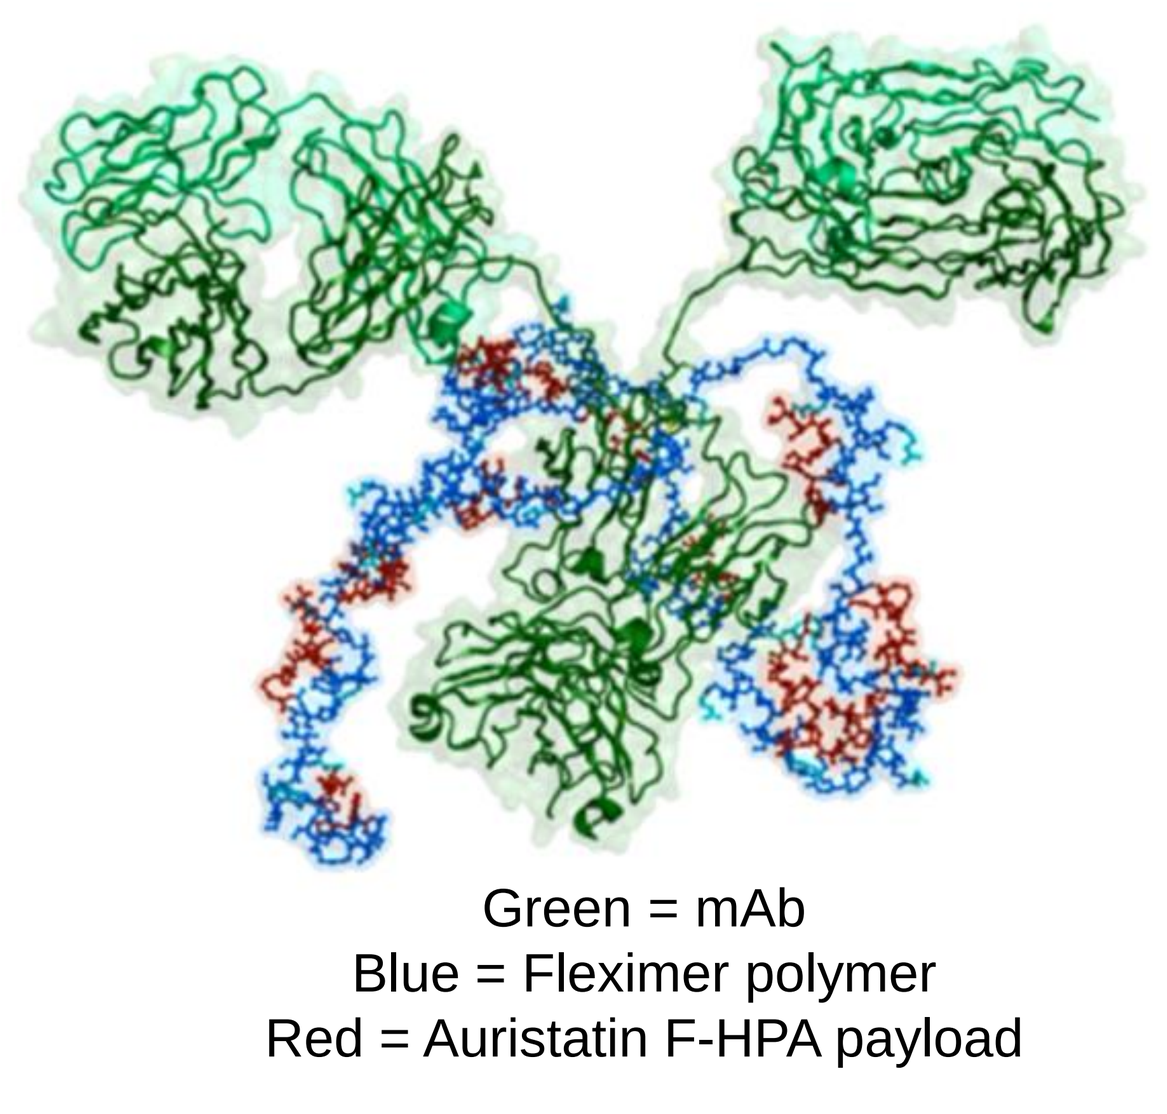

Green = mAb
Blue = Fleximer polymer
Red = Auristatin F-HPA payload

Analytes and Methods

- Conjugated Auristatin F-hydroxypropylamide (XMT-1267) was determined by extensive hydrolysis of the plasma samples followed by ESI LC MS/MS analysis on the Sciex 6500 Q-Trap (Sciex, Framingham, MA)
- Small molecule Auristatin F-hydroxypropylamide (XMT-1267), Auristatin F (XMT-1521), Monomethyl auristatin F hydroxypropyl amide (MMAF-HPA), Monomethyl auristatin F (MMAF) analysis was performed by ESI LC MS/MS Sciex 6500 Q-Trap (Sciex, Framingham, MA)
- Minor metabolite identification for excretion studies was performed using Q-Exactive Orbitrap HR ESI LC MS (ThermoFisher Scientific, Waltham, MA)
- Total XMT-1519 analysis was performed by ELISA
- LLOQ of 0.1 ng/mL was achieved for all small molecule analytes in plasma and 0.5 ng/mL in tissues

Antibody drug conjugate XMT-1522

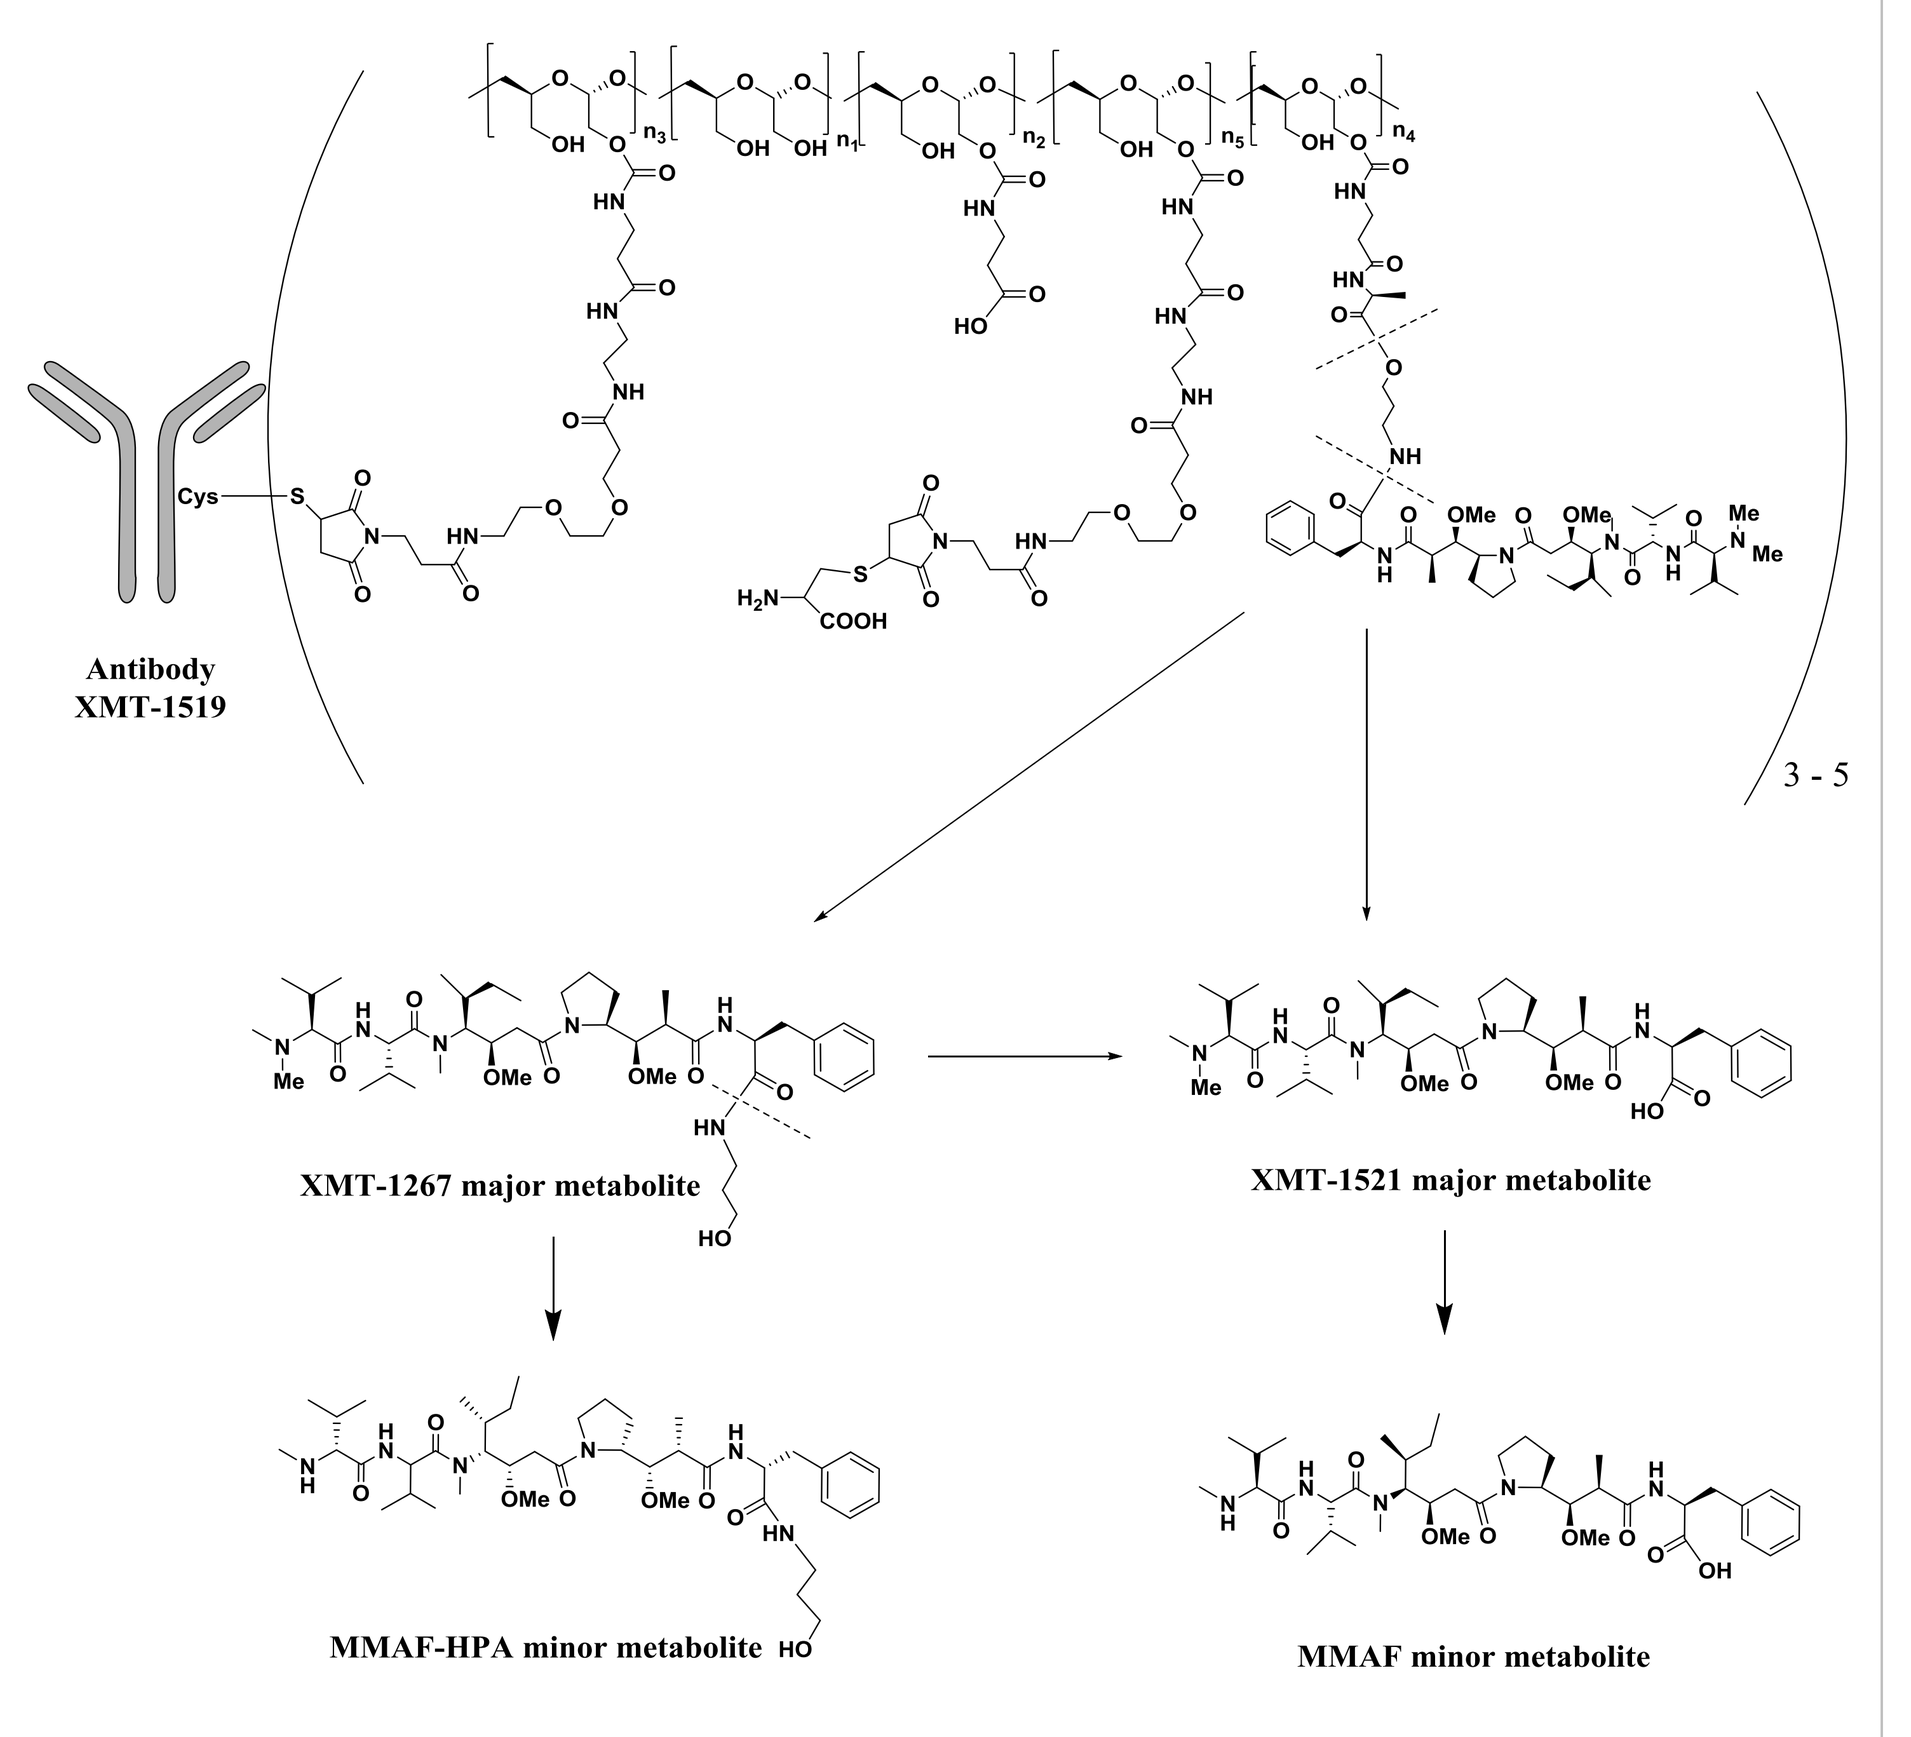
Antibody
XMT-1519
3 - 5
XMT-1267 major metabolite
XMT-1521 major metabolite
MMAF-HPA minor metabolite
MMAF minor metabolite

XMT-1522 Monkey Plasma PK Parameters

Table 1. Summary of mean XMT-1522 PK parameters for total antibody XMT-1519.

DL (mg/kg)	$T_{1/2}\beta$ (days)	T_{max} (hrs)	C_{max} (ug/mL)	$AUC_{0\text{-}inf}$ (hrs*ug/mL)	DN $AUC_{0\text{-}inf}$ (hrs*ug/mL/DL)	Vz (mL/kg)	Cl (mL/day/kg)
1.25	2.4±0.15	0.17	11.0±5.3	824±42	699±33	131.4±0.3	36.5±1.9
2.5	2.7±0.01	0.17	34.7±6.1	2411±248	964±99	100.3±0.4	25.0±2.6
5.0	3.7±0.08	0.17	104.5±18.9	6995±347	1399±69	95.4±0.4	17.0±1.0

Table 2. Summary of mean XMT-1522 PK parameters for conjugated XMT-1267.

DL (mg/kg)	$T_{1/2}\beta$ (days)	T_{max} (hrs)	C_{max} (ng/mL)	$AUC_{0\text{-}inf}$ (hrs*ug/mL)	DN $AUC_{0\text{-}inf}$ (hrs*ug/mL/DL)	Vz (mL/kg)	Cl (mL/day/kg)
1.25	2.7±0.5	0.17	1,787±722	88±2.2	70±18	113±27	27.8±0.7
2.5	2.9±0.03	0.17	4,753±484	257±7.2	103±29	83±16	19.2±0.5
5.0	3.0±0.3	0.17	9,065±1749	615±69	123±14	72±10	16.1±1.9

Table 3. Summary of mean XMT-1522 PK parameters for free XMT-1267.

DL (mg/kg)	$T_{1/2}\beta$ (days)	T_{max} (hrs)	C_{max} (ng/mL)	$AUC_{0\text{-}t}$ (hrs*ng/mL)	DN $AUC_{0\text{-}t}$ (hrs*ng/mL/DL)	t_{last} (hrs)
1.25	N/A	NA	ND	NA	NA	NA
2.5	NA	49±54	1.5±0.5	88.3±28.2	35.3±11.3	96
5.0	NA	24±19	2.8±01.3	287±221	57.3±44.4	96

Table 4. Summary of mean XMT-1522 PK parameters for free XMT-1521.

DL (mg/kg)	$T_{1/2}\beta$ (days)	T_{max} (hrs)	C_{max} (ng/mL)	$AUC_{0\text{-}t}$ (hrs*ng/mL)	DN $AUC_{0\text{-}t}$ (hrs*ng/mL/DL)	t_{last} (hrs)
1.25	N/A	N/A	N/A	N/A	N/A	N/A
2.5	N/A	84±16	0.88±0.17	77.0±4.0	31±2	168
5.0	N/A	96±0.0	1.36±0.04	226±96	45±19	336

XMT-1522 Monkey Plasma PK Profiles

- XMT-1522 PK was mostly linear, approximately dose proportional, and characterized by extended exposure to XMT-1267 drug payload, Figures 1-2.
- Clearance and volume of distribution were similar for conjugated XMT-1267 and XMT-1519 antibody, Tables 1-2.
- Exposure to free XMT-1267 and XMT-1521 was less than 1/1000th the exposure of total XMT-1267, Figure 3, Tables 3-4.
- The pharmacokinetic profiles of XMT-1522 have also been evaluated in mouse, resulting in similar outcomes
- The data indicates that the vast majority of XMT-1267 in plasma (>99.5%) is antibody-conjugated, indicating high stability of the drug polymer linkage in the systemic circulation.

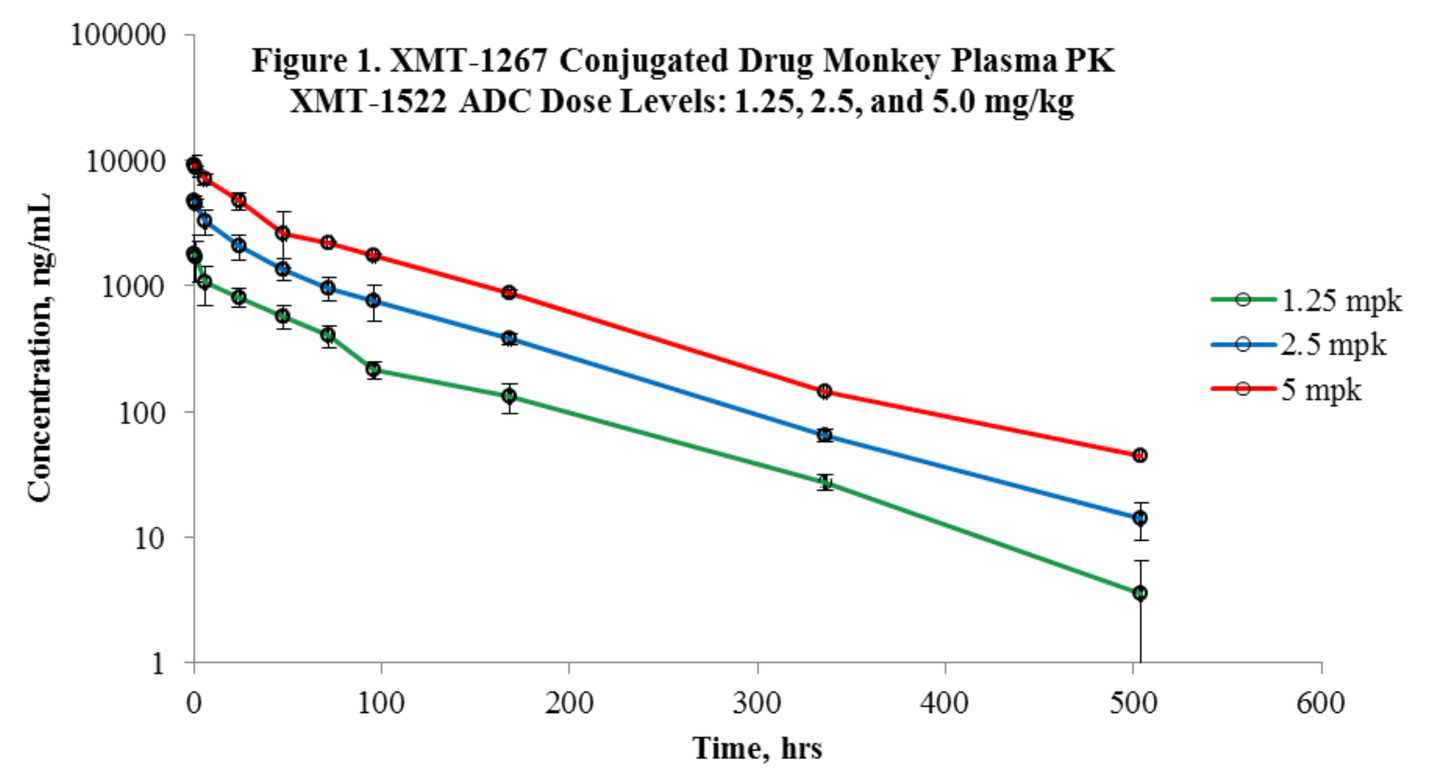
Figure 1. XMT-1267 Conjugated Drug Monkey Plasma PK
XMT-1522 ADC Dose Levels: 1.25, 2.5, and 5.0 mg/kg
Concentration, ng/mL
Time, hrs
1.25 mpk
2.5 mpk
5 mpk

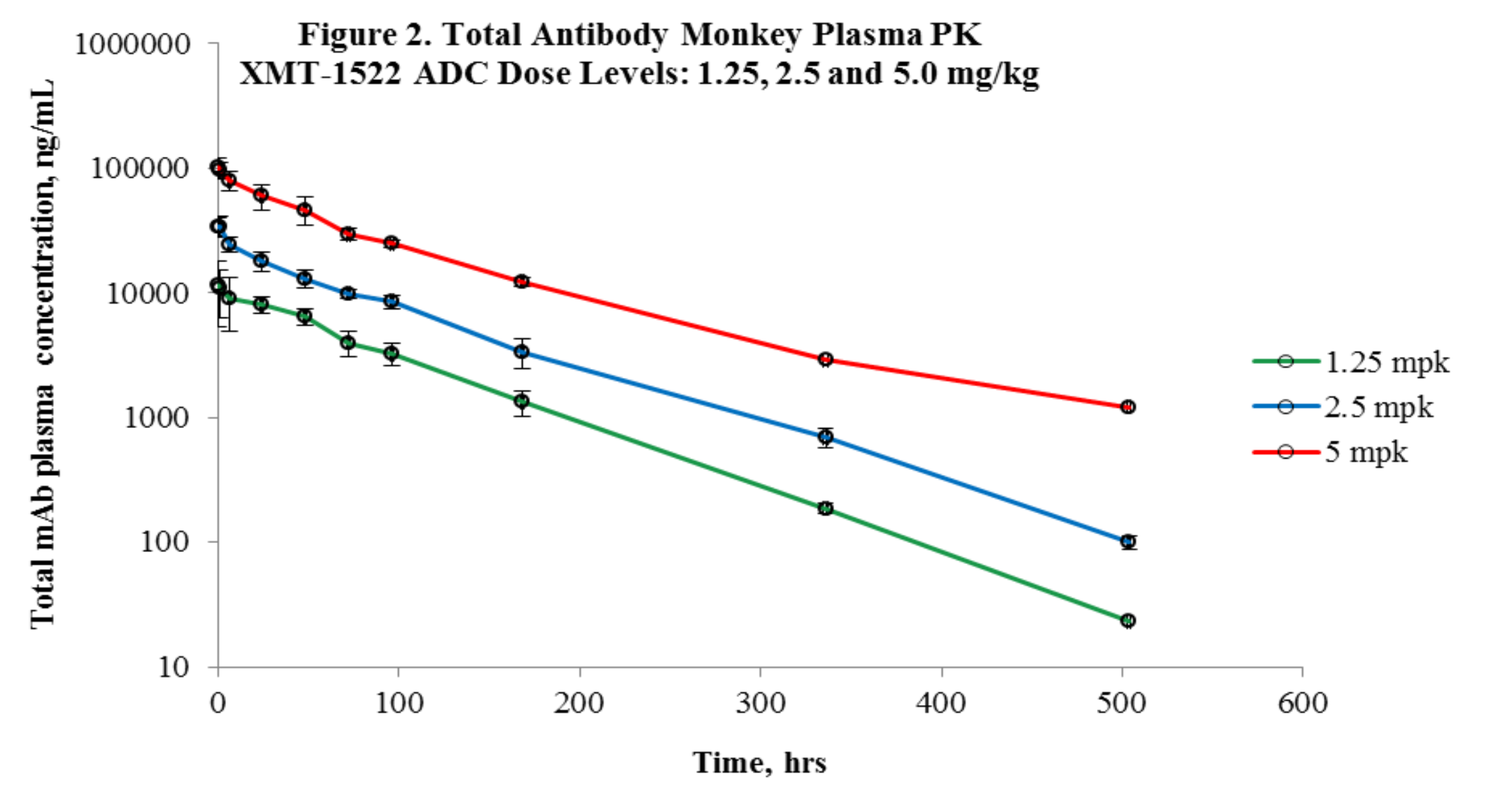
Figure 2. Total Antibody Monkey Plasma PK
XMT-1522 ADC Dose Levels: 1.25, 2.5 and 5.0 mg/kg
Total mAb plasma concentration, ng/mL
Time, hrs
1.25 mpk
2.5 mpk
5 mpk

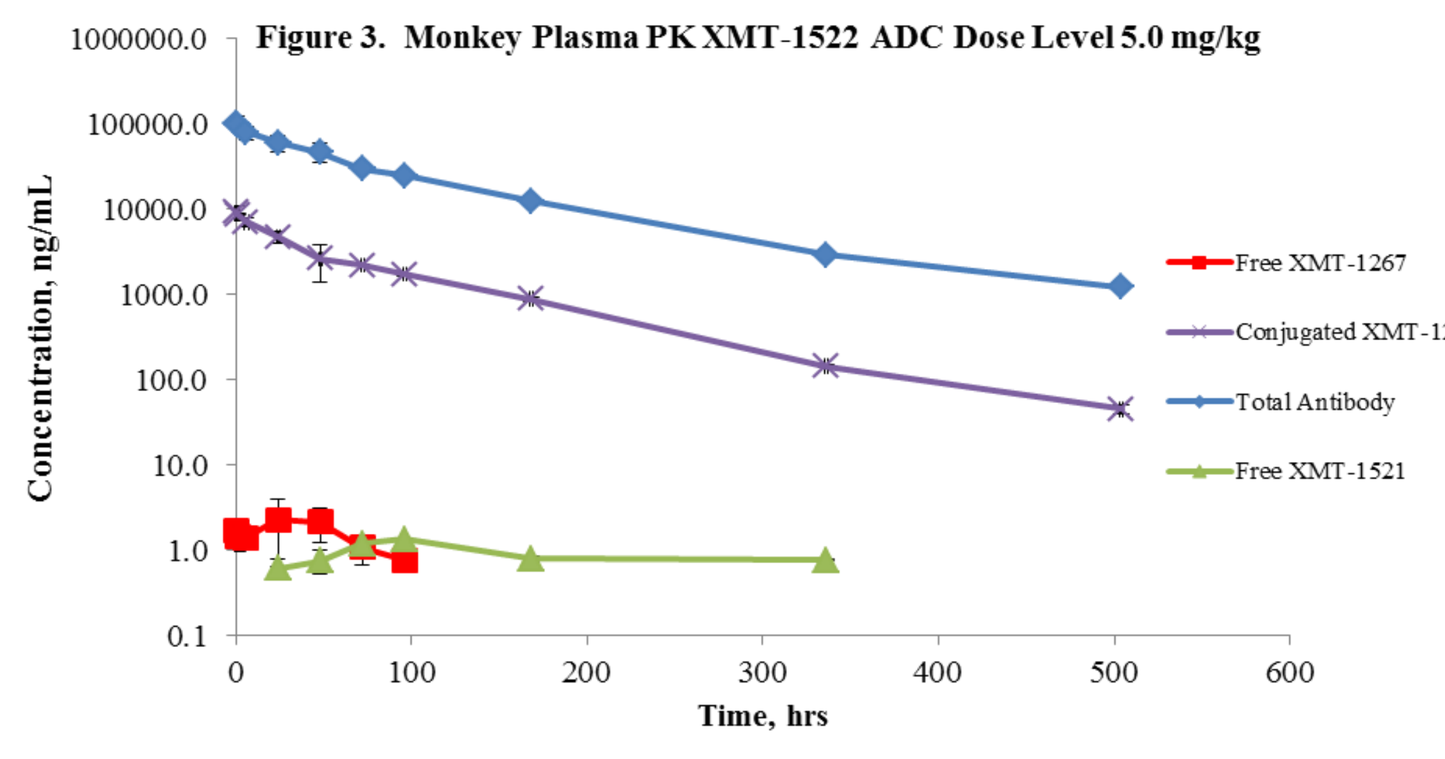
Figure 3. Monkey Plasma PK XMT-1522 ADC Dose Level 5.0 mg/kg
Concentration, ng/mL
Time, hrs
Free XMT-1267
Conjugated XMT-1267
Total Antibody
Free XMT-1521

PK/PD of XMT-1522 in the NCI-N87 Mouse Model

Plasma pharmacokinetics and tissue disposition of XMT-1522 related compounds after a single intravenous administration of XMT-1522 (3.0 mg/kg dose level) in the NCI-N87 human gastric carcinoma tumor xenograft model using female BC.17 SCID mice.

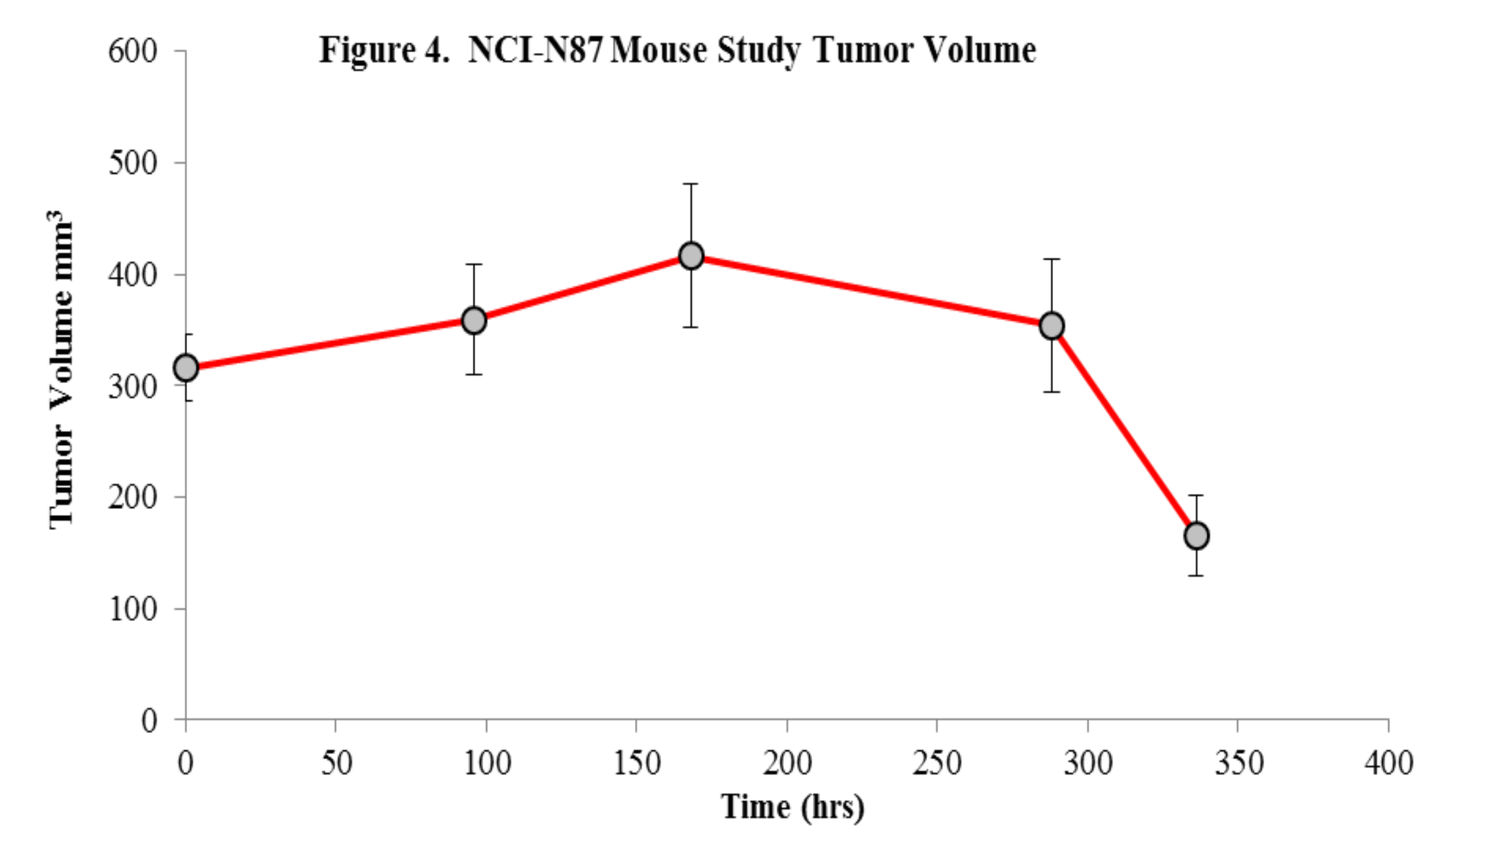
Figure 4. NCI-N87 Mouse Study Tumor Volume
Tumor Volume mm³
Time (hrs)

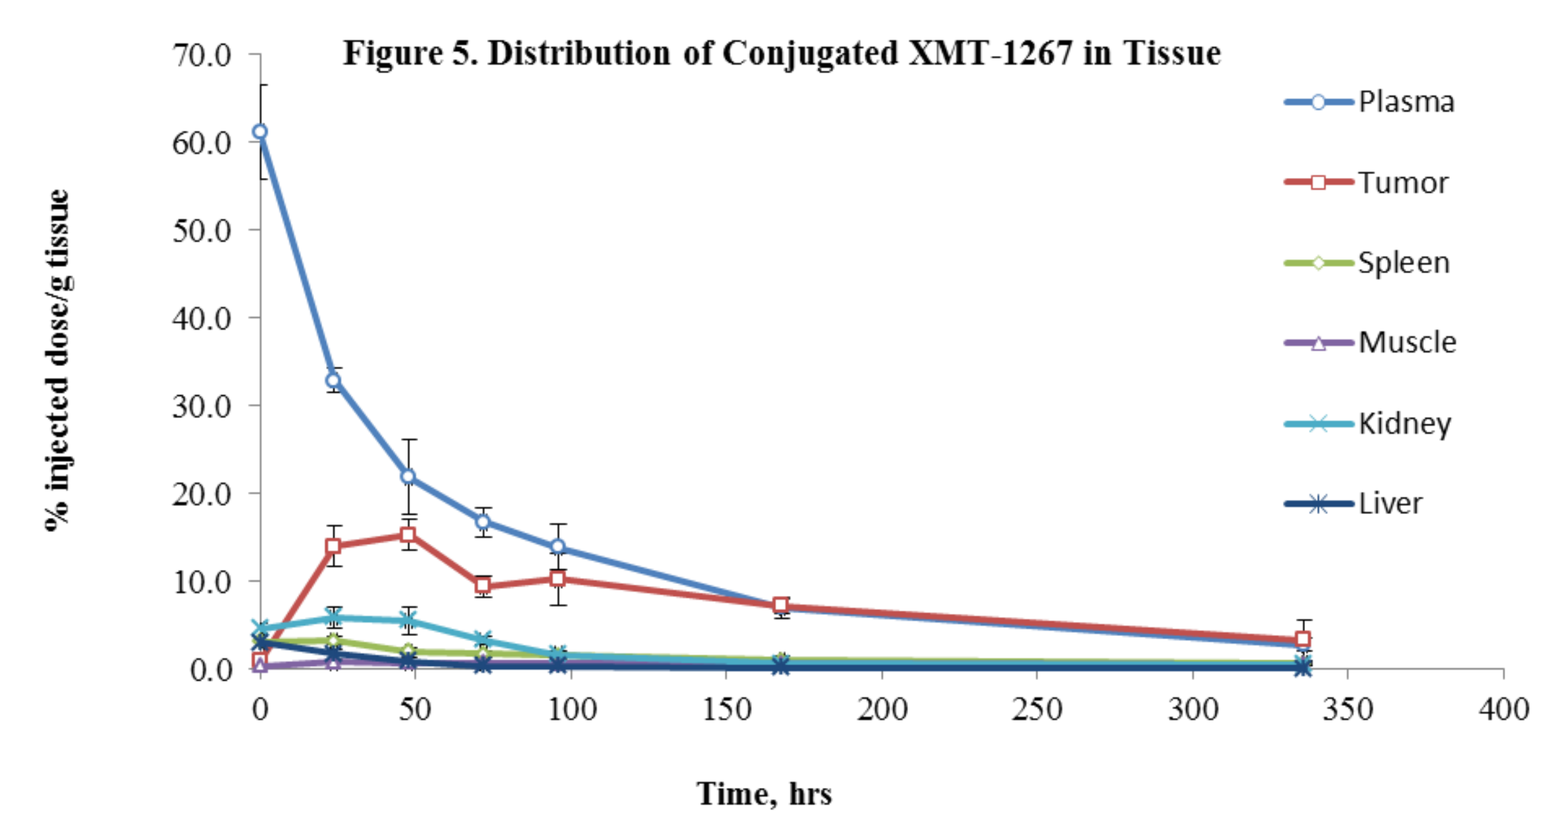
Figure 5. Distribution of Conjugated XMT-1267 in Tissue
% injected dose/g tissue
Time, hrs
Plasma
Tumor
Spleen
Muscle
Kidney
Liver

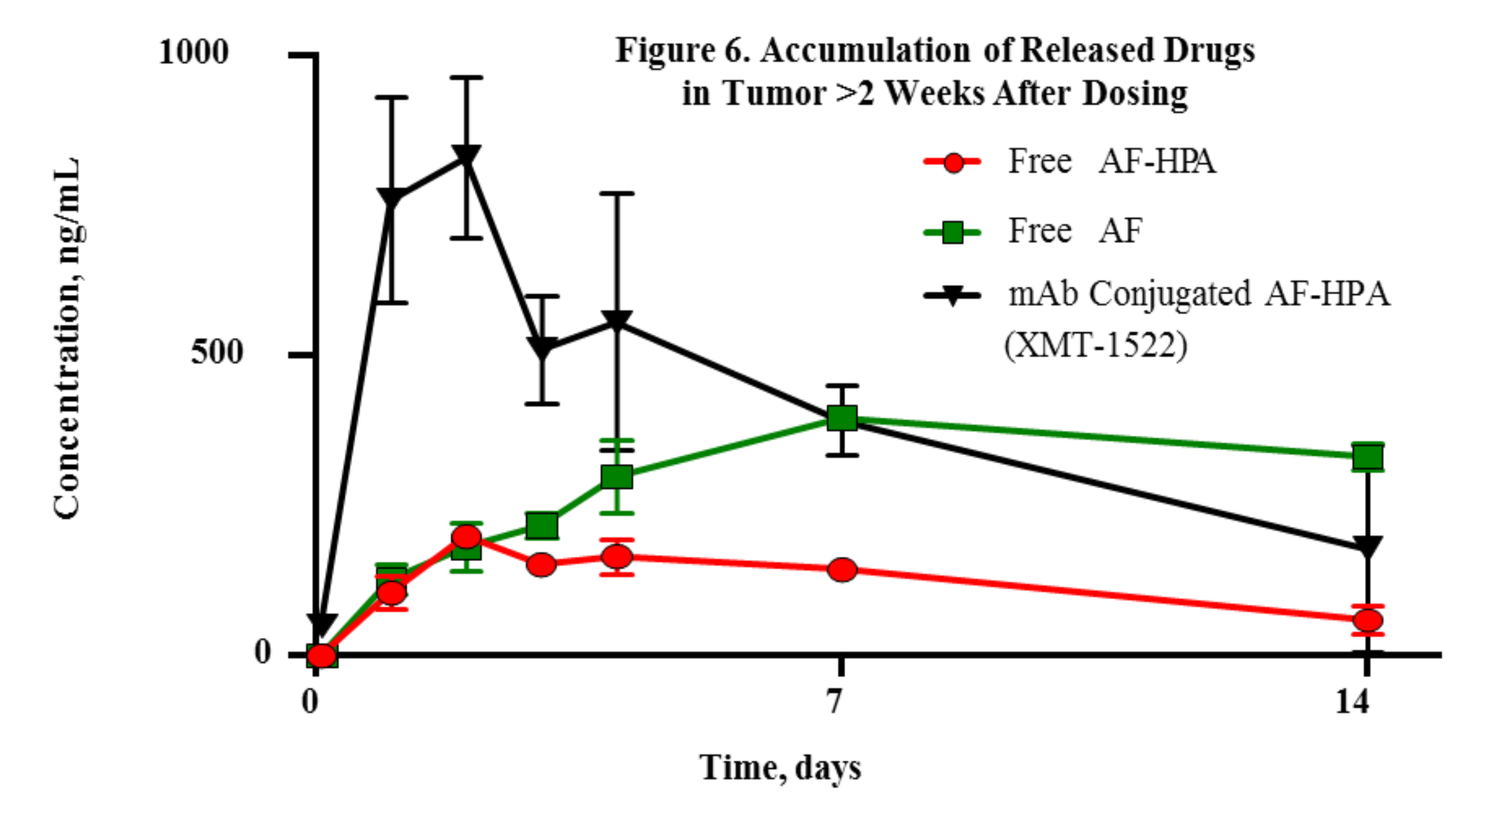
Figure 6. Accumulation of Released Drugs in Tumor >2 Weeks After Dosing
Concentration, ng/mL
Time, days
Free AF-HPA
Free AF
mAb Conjugated AF-HPA (XMT-1522)

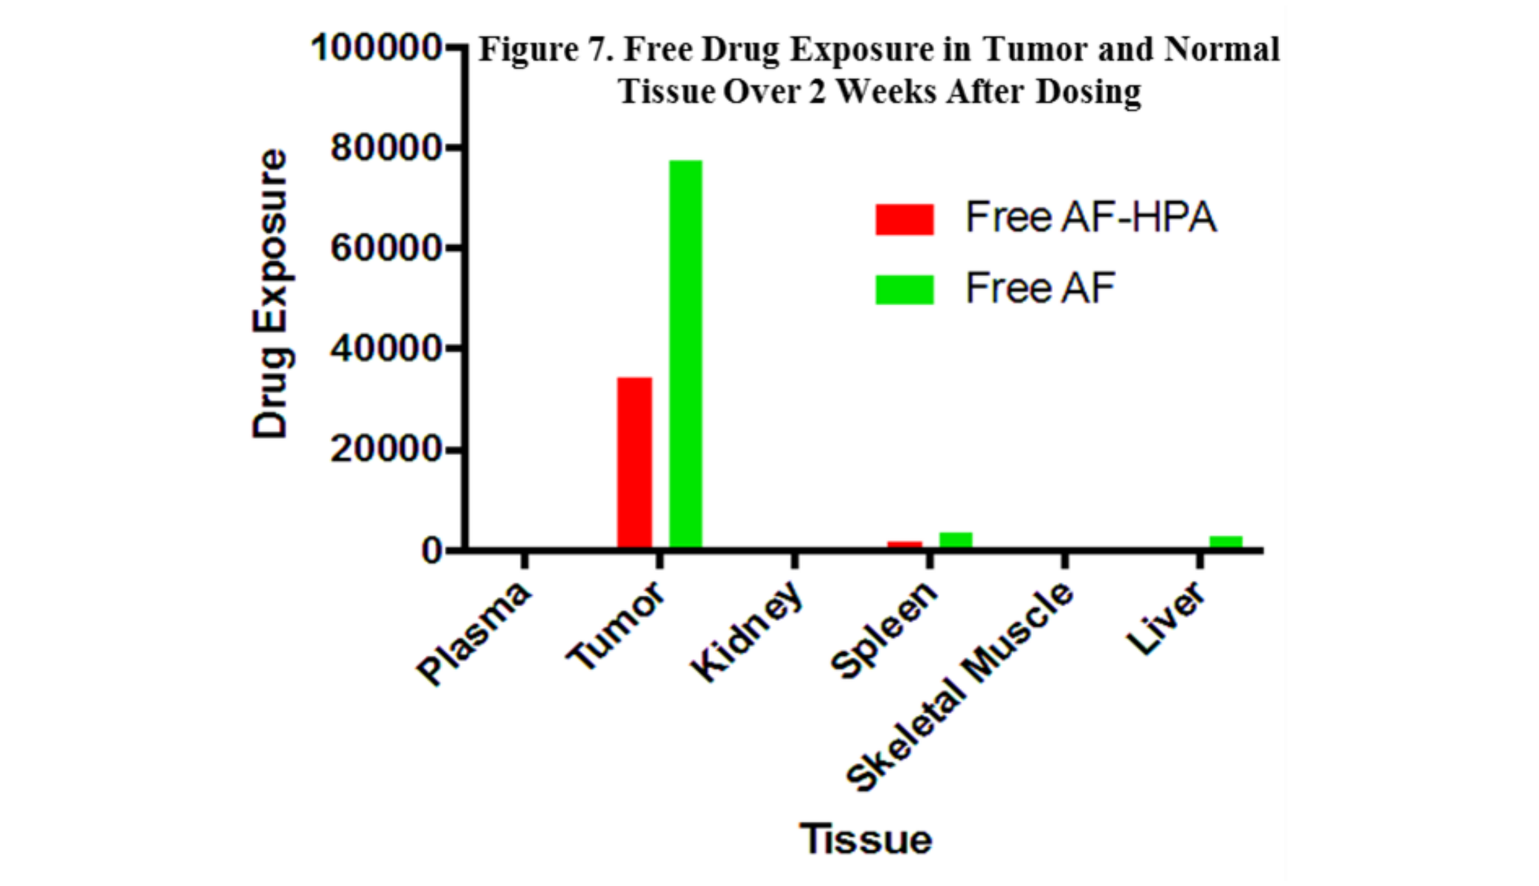
Figure 7. Free Drug Exposure in Tumor and Normal Tissue Over 2 Weeks After Dosing
Drug Exposure
Free AF-HPA
Free AF
Plasma
Tumor
Kidney
Spleen
Skeletal Muscle
Liver
Tissue

Elimination of XMT-1522 Related Compounds

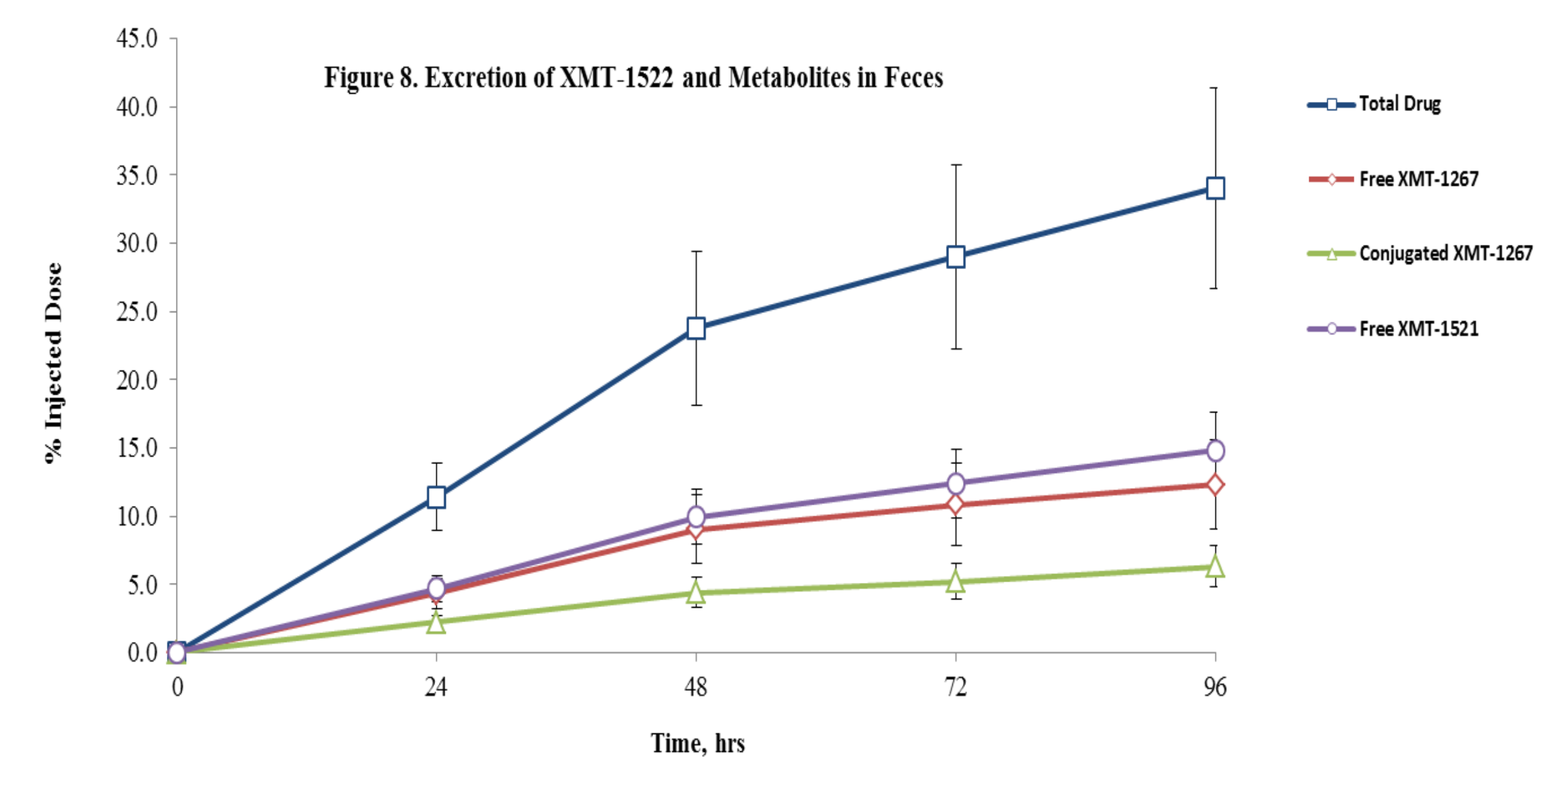
Figure 8. Excretion of XMT-1522 and Metabolites in Feces
% Injected Dose
Time, hrs
Total Drug
Free XMT-1267
Conjugated XMT-1267
Free XMT-1521

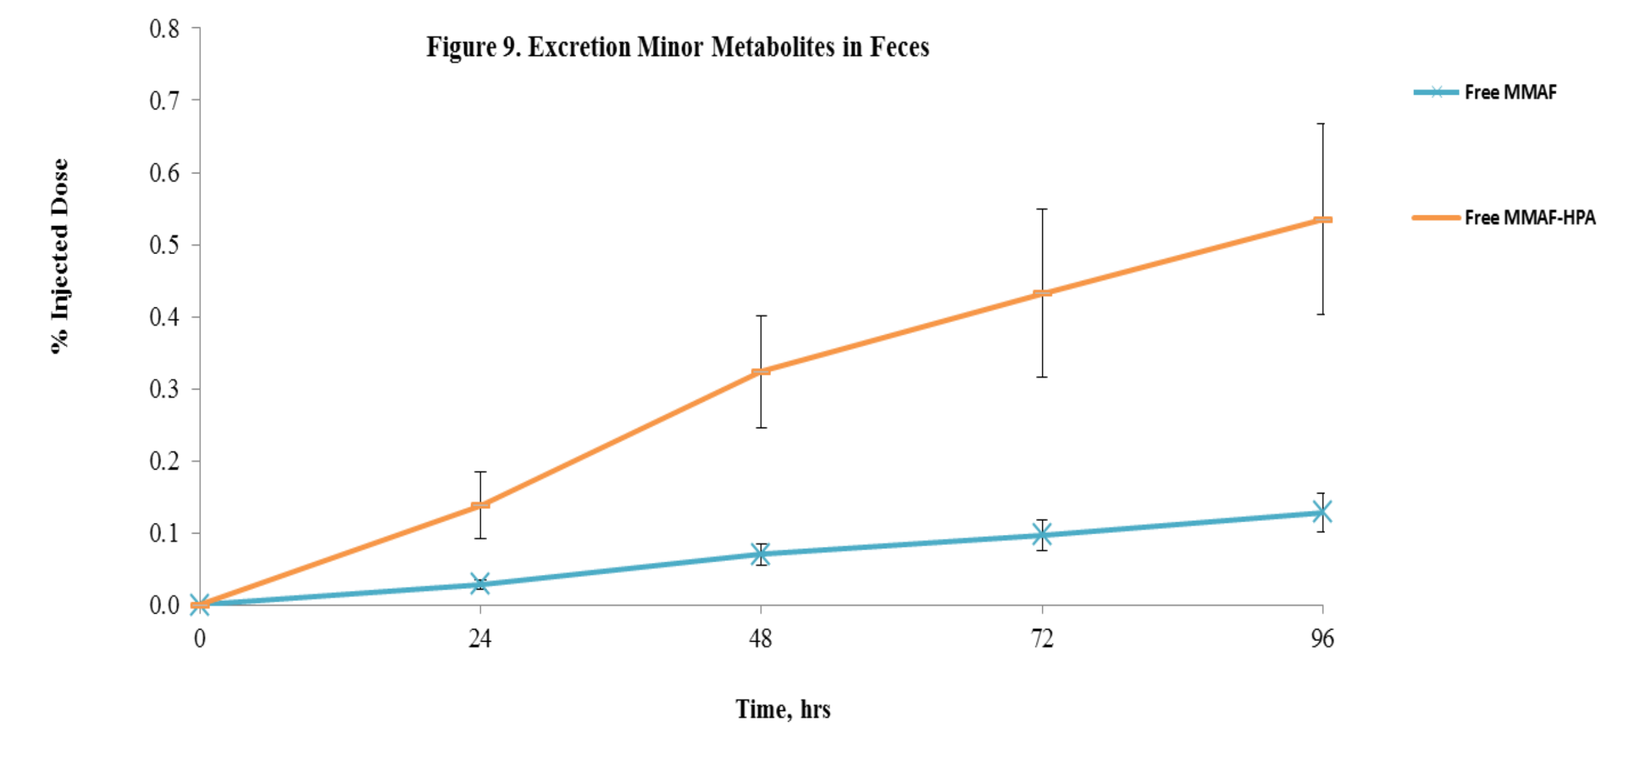
Figure 9. Excretion Minor Metabolites in Feces
% Injected Dose
Time, hrs
Free MMAF
Free MMAF-HPA

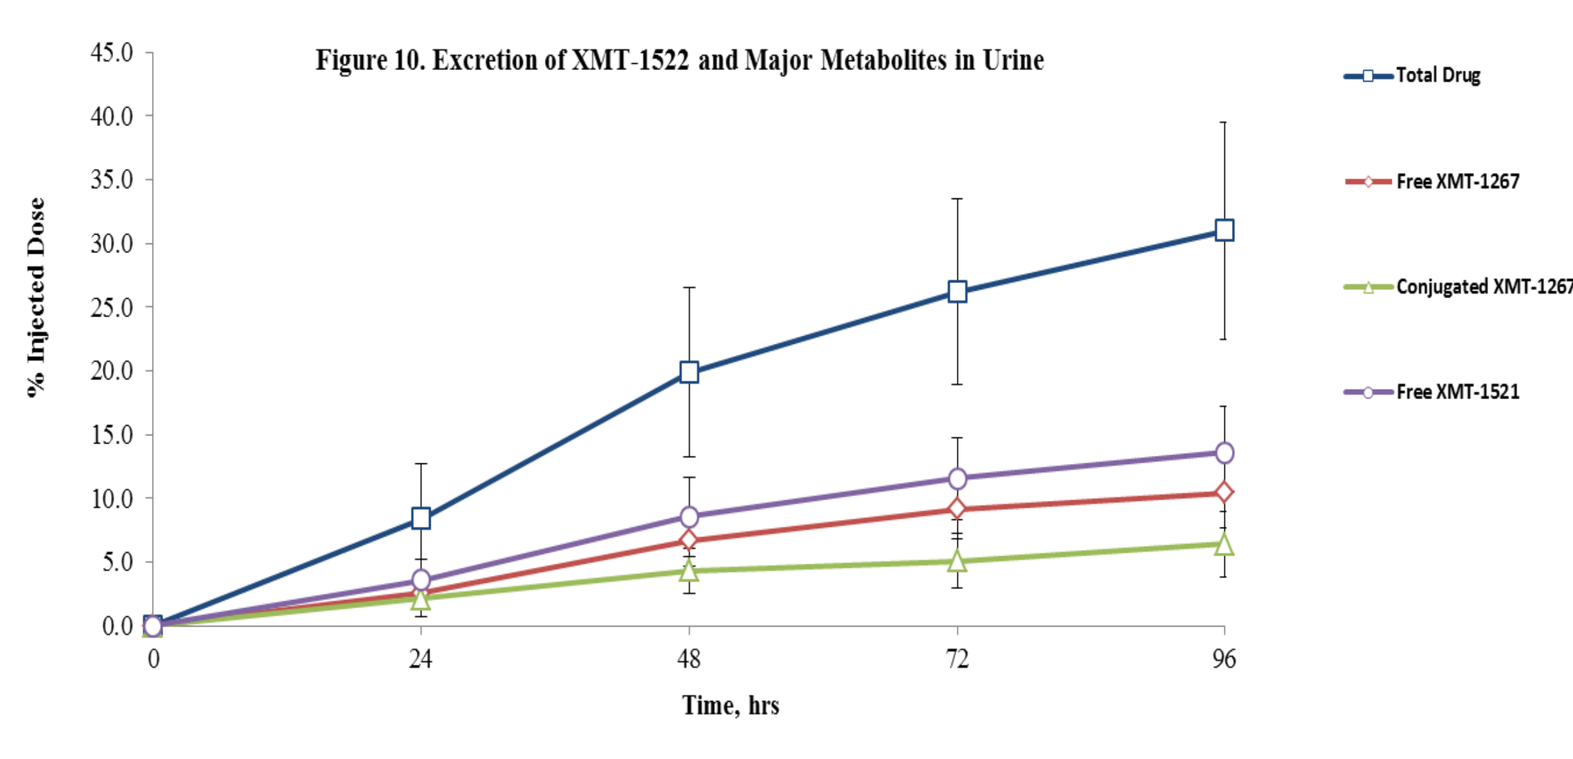
Figure 10. Excretion of XMT-1522 and Major Metabolites in Urine
% Injected Dose
Time, hrs
Total Drug
Free XMT-1267
Conjugated XMT-1267
Free XMT-1521

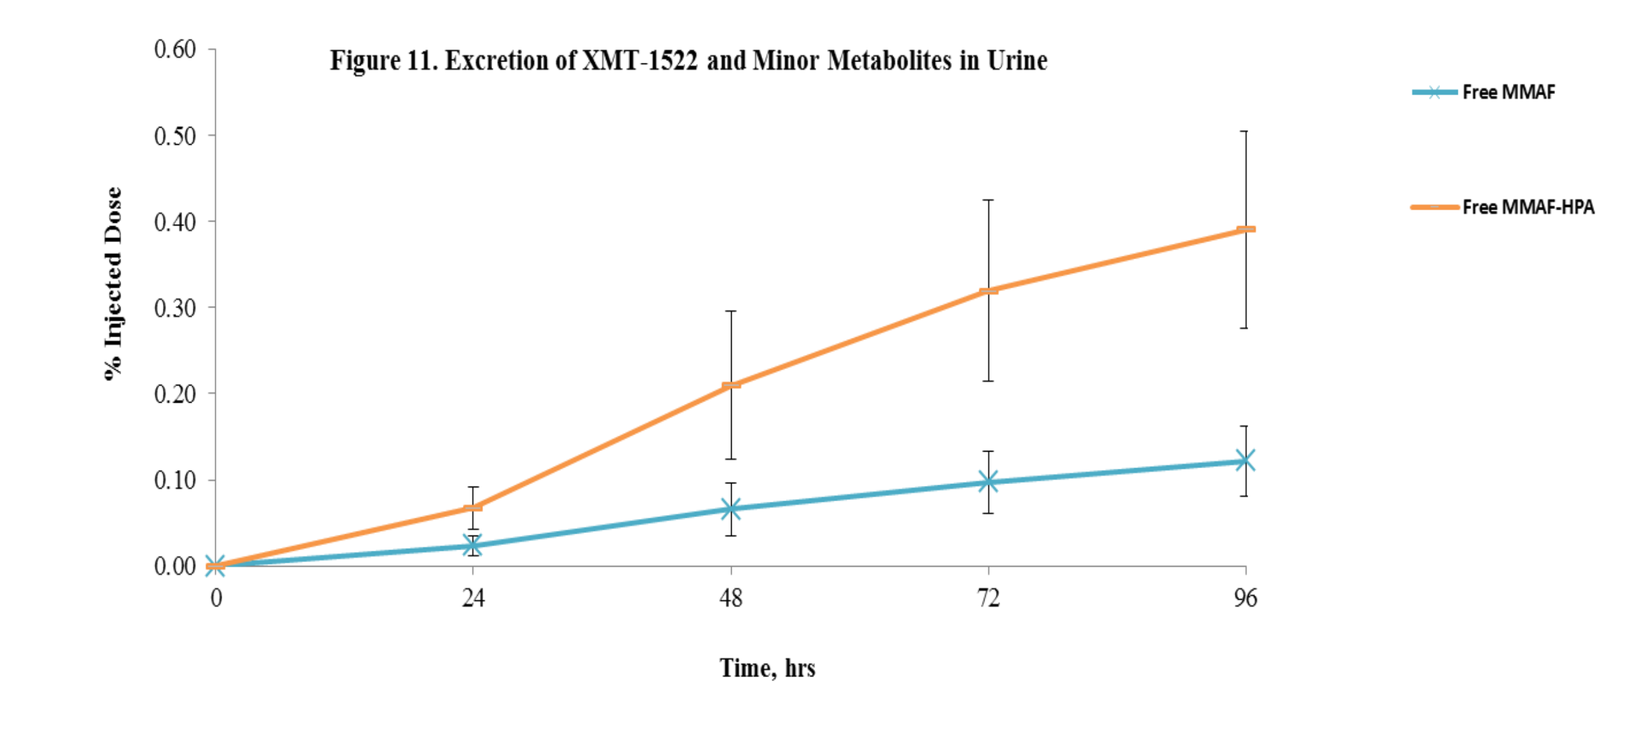
Figure 11. Excretion of XMT-1522 and Minor Metabolites in Urine
% Injected Dose
Time, hrs
Free MMAF
Free MMAF-HPA

In Vitro Plasma Stability of XMT-1522

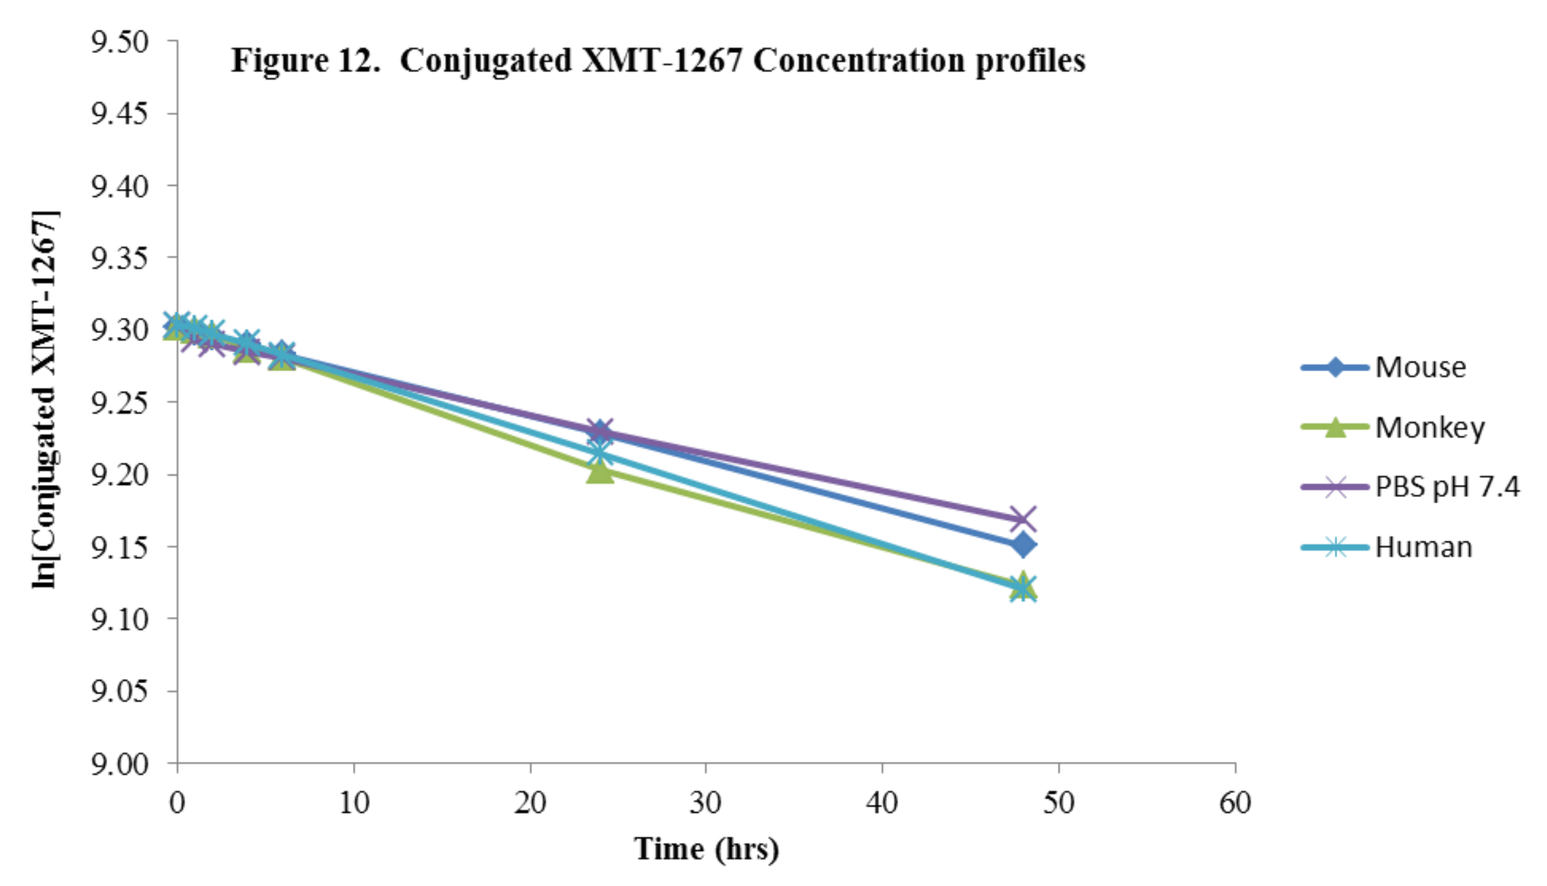
Figure 12. Conjugated XMT-1267 Concentration profiles
ln[Conjugated XMT-1267]
Time (hrs)
Mouse
Monkey
PBS pH 7.4
Human

- The LC/MS/MS analysis of XMT-1522 plasma incubations has shown the presence of two XMT-1522 drug release products: XMT-1267 and XMT-1521. The results suggest that XMT-1521 is generated mostly by enzymatic processing of ADC release product XMT-1267 via proteolytic cleavage (amidolysis) of carboxy-terminal hydroxypropyl amido group of XMT-1267
- XMT-1522 ADC was significantly (60 to 200 times) more stable in animal plasma *in vitro* than the structurally related small molecule ester, XMT-1864, which indicates significant contribution of XMT-1522 polymer based drug linker microenvironment to both enzymatic and hydrolytic stability of drug conjugate linkage.

Table 5. Conjugated drug XMT-1267 animal plasma half-life *in vitro* (37°C, pH 7.4)

Matrix	XMT-1522 T ½ (hrs) Active plasma and PBS	XMT-1864 T ½ (hrs) Active plasma and PBS
Mouse	224	<1.0
Monkey	217	2.1
Human	182	3.0
PBS pH 7.4	182	5.6

Summary and Conclusions

Cyno Monkey PK Profile:

● Almost identical monkey plasma PK profiles for total antibody and conjugated drug indicated stability of XMT-1522 in circulation and slow rate of drug release. No significant change in conjugated drug to antibody plasma concentration ratio between T_0 and the last sampling point (504 hrs) indicated stability of both drug/polymer and polymer/antibody linkers.

● The XMT-1267 plasma concentration was significantly lower (> 1,000 times) than plasma concentration of the parent conjugated drug. After XMT-1522 administration, XMT-1267 was detectable in plasma for one week, reaching Cmax at approximately 24 hours post administration. The delayed XMT-1267 Cmax can be attributed to intracellular enzymatic processing of the ester-based drug linker resulting from both target-specific and non-targeted ADC tissue uptake. Similar to conjugated drug, the exposure to free XMT-1267 was slightly more than dose proportional.

● The major drug metabolite XMT-1521 had delayed plasma appearance relative to parent XMT-1267 (Tmax 96 hours) and was detectable in plasma for up to 2 weeks (see Figure 3 and Table 4). The plasma exposure to XMT-1521 was comparable to the level of parent XMT-1267 exposure but more than 1000 times lower than exposure to conjugated XMT-1267. The XMT-1521 plasma exposure was slightly more than dose proportional.

Mouse Tissue Distribution, NCI N-87 Tumor Bearing Mouse Model:

● In all tissues conjugated XMT-1267 was the major XMT-1522 drug related compound contributing to tissue exposure. Maximum tissue exposure to conjugated XMT-1267 was observed for plasma (>99% total drug exposure AUC0-t last).

● Plasma exposure AUC0-t last to conjugated XMT-1267 over 336-hour time period was approximately 3,000 times higher than exposure AUC0-t last to small molecule drug XMT-1267. In contrast, tumor exposure to both free XMT-1267 and XMT-1521 was significant. Cumulatively, XMT-1267 and XMT-1521 contributed to approximately 50% of total drug exposure in tumor. Overall, tumor total drug exposure AUC0-t last was approximately 23% higher than respective plasma total drug exposure, indicating tumor specific accumulation of XMT-1522.

● Despite the high kidney tissue concentration for conjugated XMT-1267 (Cmax 257 ng/g tissue), no significant kidney tissue concentration was observed for free XMT-1267 or XMT-1521 at any time point. After reaching maximum XMT-1267 and XMT-1521 kidney accumulation at 24 hours (Cmax 0.9 ng/g and 4.4 ng/g, respectively), the concentrations of XMT-1267 and XMT-1521 rapidly declined, resulting in both metabolites being not detectable in kidney at time points later than 72 hours post administration. Cumulative kidney tissue exposure AUC0-t last to metabolites XMT-1267 and XMT-1521 was low, accounting for approximately 1.3 % of total drug tissue exposure.

● Tissue analysis indicated that both XMT-1267 and its metabolite XMT-1521 were generated intracellularly from XMT-1522, and that the carboxylate-containing active metabolite XMT-1521 was retained in tumor tissue over 2 weeks, suggesting intracellular trapping.

Rat Excretion Studies:

● XMT-1522 excretion studies, conducted in rat, indicated that the XMT-1267 payload was mainly excreted by the gastrointestinal route. In the first 96 hours after administration 33% of the XMT-1267 dose was excreted in feces, compared to 3% excreted in urine. The major contributing metabolites both in feces and urine were conjugated XMT-1267, XMT-1521, and free XMT-1267.

Conclusion:

● XMT-1522 ADC has plasma kinetics, tissue distribution and excretion profile favorable for clinical evaluation and development.

Footer for e-pub or distribution: AACR, April 2017